RARE ISN'T RARE

Rare cancers compose up to 50% of all U.S. diagnoses

SHEPHERD THERAPEUTICS

Katherine Arline, Robert Louis Treuting, Rodrigo Bravo, Emily Zeme, Jamie Dempsey Barber, William Siders, Johanne Kaplan

Poster 1198

OBJECTIVES

I. To understand the incidence of rare cancers in the United States

II. To identify and discuss clinical trial availability for rare and non-rare cancer patients

III. To highlight the translation gap in targeted therapies for rare and non-rare cancers

INTRODUCTION

Rare cancers are an understudied and deadly public health concern. Estimates for the percentage of cancer diagnoses that carry the "rare" nomenclature vary depending upon the source cited and the statistical definition of what constitutes a rare cancer. Among the dearth of literature focused on rare cancer, the most-repeated metric states that about one quarter of all adult cancers are rare. Given the evolving landscape of cancer biology and advances in disease classification and diagnoses, we argue that oft-cited percentages can underestimate the burden of rare cancers, especially from the perspective of neglected patient populations and a lack of translational research. As such, we have found that rare cancer in total is likely underestimated as a proportion of cancer diagnoses, and that those cancers disproportionately affect historically underserved populations including women and children. In addition, treatment options are needed for each cancer subtype and should be developed based on a thorough understanding of each cancer's molecular biology. Finally, we discuss the implications behind a lack of proportionate clinical trial coverage, which contributes to a gap in available targeted therapies.

DATA HIGHLIGHTS

- **Fig. 1: According to ACS metrics, 29.70% of cancer diagnoses are rare; NIH metrics put this number at 50.52%.**
 - Additional analysis, not displayed, shows that of 52 major groupings of cancer, such as the broad classification of breast cancer, 35 (67.3%) disproportionately affect minorities in terms of incidence or overall survival.
 - Seven groups exclusively or almost exclusively affect women, and 7 additional groups display disproportionate incidence or negative outcomes in women.
 - While many cancers affect children, and while all cancer in children is rare, at least 55 cancers exclusively or frequently affect children.
- **Fig. 2: An overwhelming 74.89% of clinical trials in Phases 1-3 did not include a rare cancer between January 1, 2012 and January 1, 2017.**
 - Fig. 2E: In each phase of clinical trials, rare cancers were underrepresented in a statistically significant manner relative to the proportion of cancers that are rare:
 - The proportion of phase I clinical trials that considered at least one rare cancer (M = 0.237, SD = 0.425) compared to the overall proportion of cancers that are considered rare, t(1798) = -6.304, p= 1.82e-10).
 - The proportion of phase II clinical trials that considered at least one rare cancer (M = 0.279, SD = 0.448) compared to the overall proportion of cancers that are considered rare, t(1793) = -2.011, p = 0.02224).
 - The proportion of phase II clinical trials that considered at least one rare cancer (M = 0.195, SD = 0.396) compared to the overall proportion of cancers that are considered rare, t(400) = -5.3299, p = 0.02224).
- **Fig. 3: Fig 3A: Every cancer that did not receive any phase 3 development between January 1, 2012 and January 1, 2017 was a rare cancer.** In 2017, this translated to 113,962 new patients whose cancers had not been included in any phase 3 trials in the preceding five years. In that period, all non-rare cancers were included in phase 3 clinical trials.
 - Fig. 3B: Less than 20% of the money dedicated to all phase 3 clinical trials were committed to trials including a rare cancer.
- **Fig. 4: 83.23% of all patients who lack an FDA-approved targeted therapy are rare cancer patients.**

METHODOLOGY & SOURCES

Fig. 1: We deconstructed the most recent publicly available epidemiological data regarding the incidence of all cancers in the United States, using the two most cited definitional metrics:

- Following the National Cancer Institute (NCI) definition of a rare cancer as affecting < 15 persons per 100,000 per year, for 2017, a cancer affecting 45,691 or fewer was classified as rare.
- Following the Rare Diseases Act of 2002 and metric used by the American Cancer Society (ACS) defining a rare cancer as affecting < 6 persons per 100,000 per year, for 2017, a cancer affecting 19,584 or fewer was classified as rare.

We carefully considered each broad cancer type and characterized subtypes using criteria derived from distinctions found in the literature (in order of most to least influential variable): SEER and ACR's databases' categorical organizations, location of affliction in the human body, curriculum in oncology training, and approved therapeutic methods.

We collected incidence data primarily from ACS and SEER, and cited literary or patient-focused sources if rare cancer subtypes were not found in either primary databases.

These numbers include estimated new cases in 2017, and incidence rates. These rates are age-adjusted and based on 2010-2014 cases and deaths. For the American Cancer Society (ACS) estimates, the new cases were estimated based on 1999-2013 incidence data reported by the North American Association of Central Cancer registries (NAACCR). These methods resulted in a total cancer count of 1,692,163, which is less than 1% removed from SEER's 2017 estimated cancer incidence.

Fig. 2: To access clinical trial data, on clinicaltrials.gov:

- The term "cancer" was entered into the "Condition or Disease" field, no additional information was added to the "Other terms" field, the term "interventional Studies (Clinical Trials)" was selected for the Study type field, the term "All Studies" was selected for the Study Results field, all Clinical studies were selected for the Recruitment status field (this includes Not yet recruiting, Recruiting, Enrolling by invitation, Active, not recruiting, Suspended, Terminated, Completed, Withdrawn, and Unknown status).
- Under Eligibility Criteria: all age groups were selected for the Age Group field, all were selected for the Sex field.
- Under Targeted search: no additional information was added to the Intervention/treatment field, no additional information was added to the Title / Acronym field, no additional information was added to the Outcome Measure field, no additional information was added to the Sponsor / Collaborator field, no additional information was added to the Sponsor (Lead) field, and no additional information was added to the Study IDs field.
- Under Locations: the term "United States" was selected for the Country field (the fields State, City, and Distances were left blank), and no additional information was added to the Location Terms field.
- Under Additional Criteria: either Phase 1, Phase 2, or Phase 3 was selected for the Phase field, the term "Industry" was selected for the Funder Type field, and the date "1/1/2012" (MM/DD/YYYY) was select for the From field and the date "1/1/2017"(MM/DD/YYYY) was select for the Start field under the Study Start field (thus, showing clinical trial studies starting between the 5-year span of 1/1/2012 to 1/1/2017), and no additional information was added to the remaining fields of Primary Completion, First Posted, and Last Update Posted.
- With all these search Criteria filled, the search was applied to clinicaltrials.gov; results were downloaded, where all the Found Studies were selected for the Number of Studies field, "all Available Columns" were selected for the Select table columns field, and "Comma-separated values" were selected for the Select file format field.
- We downloaded results to a specific .csv file saved under a unique name (e.g. CT_Phase1_original). A modified version was then created, removing certain columns and adding three new columns in order to analyze which trials' studies included rare cancers, non-rare cancers, a mix of rare and non-rare cancers, and Hematological cancers. The two previously mentioned metrics for rare (<6/100,000) and (<15/100,000) were used to parse the rare cancer data into two categories, across a 5 year span from 2012 to 2017.

Of the clinical trials studied in SHEPHERD's report, there were 426 phase 1 trials that included a rare cancer, 500 phase 2 trials that included a rare cancer, and 78 phase 3 trials that included at least one rare cancer. In contrast, there were 1373 phase 1, 1294 phase 2, and 323 phase 3 trials that did not study a rare cancer. For each phase, we considered each trial to be a Bernoulli random variable that was either 0 or 1, with 0 corresponding to the trial not including a rare cancer and 1 corresponding to the trial including at least one rare cancer. This allowed us to calculate the mean and the variance of the proportion of trials for each phase that included at least one rare cancer. We then performed a one sample t-test for each trial, with the null hypothesis being that the proportion of trials including at least one rare cancer is aligned with the proportion of cancers that are considered rare. Finally, the p-values from the t-test were adjusted using the Holm-Bonferroni method to account for the fact that we performed three t-tests.

Fig. 3: Data was extrapolated from the clinical trials analysis and combined with assessed incidence data. To avoid overcounting, large indications were divided into rare and non-rare populations.

Fig. 4: We calculated the number of cases for 2017 in our proprietary cancer list using the ACS definitional metric of rare. Then, we identified indications with at least one form of FDA-targeted therapy. Using the 2017 incidence data, we assessed that patient population's access to an FDA targeted therapy for both the rare and non rare patient populations.

FIGURE 1: INCIDENCE OF RARE CANCERS IN THE UNITED STATES, 2017

	NCI <15/100,000 Rare Definition	ACS <6/100,000 Rare Definition
Total cancers	396	396
Total numbers of rare cancers	386 (97.47%)	374 (94.44%)
Total number of non-rare cancers	10 (2.53%)	22 (5.56%)
New rare cancer patients	854,849 (50.52%)	502,636 (29.70%)
New non-rare cancer patients	837,314 (49.48%)	1,189,527 (70.3%)

2017 Incidence/year is the predicted number of new cases of that indication for the entire year of 2017. Working US population: 326,395,657 (accessed 12/7/2017 at 5:38PM EST from the U.S. Census Bureau). Rare for <6/100,000: One metric to determine rare cancer is if there is less than 6 cases in a population of 100,000 per year, or <6/100,000. This roughly translates into <19,583.73 cases per year in the US. Rare for <15/100,000: Another metric to determine rare cancer is if there are less than 15 cases in a population of 100,000 per year, or <15/100,000. This roughly translates into <48,959.35 cases per year in the US. These rates are age-adjusted and based on 2010-2014 cases and deaths. For the American Cancer Society (ACS) estimates, the new cases were estimated based on 1999-2013 incidence data reported by the North American Association of Central Cancer registries (NAACCR).

FIGURE 2: CLINICAL TRIALS AVAILABLE TO RARE CANCER PATIENTS 2012-2016

FIGURE 2A: PHASE 1 CLINICAL TRIALS, 2012-2016

	<15/100,000 Rare Definition	<6/100,000 Rare Definition
Rare cancer only trials	523 (29.07%)	192 (10.67%)
Non-rare cancer trials	1038 (57.7%)	1373 (76.32%)
Mixed trials that include at least 1 rare cancer	238 (13.23%)	234 (13.01%)

FIGURE 2B: PHASE 2 CLINICAL TRIALS, 2012-2016

	<15/100,000 Rare Definition	<6/100,000 Rare Definition
Rare cancer only trials	7802 (44.71%)	363 (20.23%)
Non-rare cancer trials	889 (49.55%)	1294 (72.13%)
Mixed trials that include at least 1 rare cancer	103 (5.74%)	137 (7.66%)

FIGURE 2C: PHASE 3 CLINICAL TRIALS, 2012-2016

	<15/100,000 Rare Definition	<6/100,000 Rare Definition
Rare cancer only trials	168 (41.9%)	59 (17.71%)
Non-rare cancer trials	220 (55.86%)	323 (80.55%)
Mixed trials that include at least 1 rare cancer	13 (3.24%)	19 (4.74%)

FIGURE 2D: PHASE 1-3 CLINICAL TRIALS, 2012-2016

	<15/100,000 Rare Definition	<6/100,000 Rare Definition
Rare cancer only trials	1493 (37.38%)	614 (15.37%)
Non-rare cancer trials	2147 (53.76%)	2990 (74.89%)
Mixed trials that include at least 1 rare cancer	354 (8.86%)	390 (9.77%)

FIGURE 2E: RARE AND NON-RARE CANCER CLINICAL TRIALS, 2012-2016

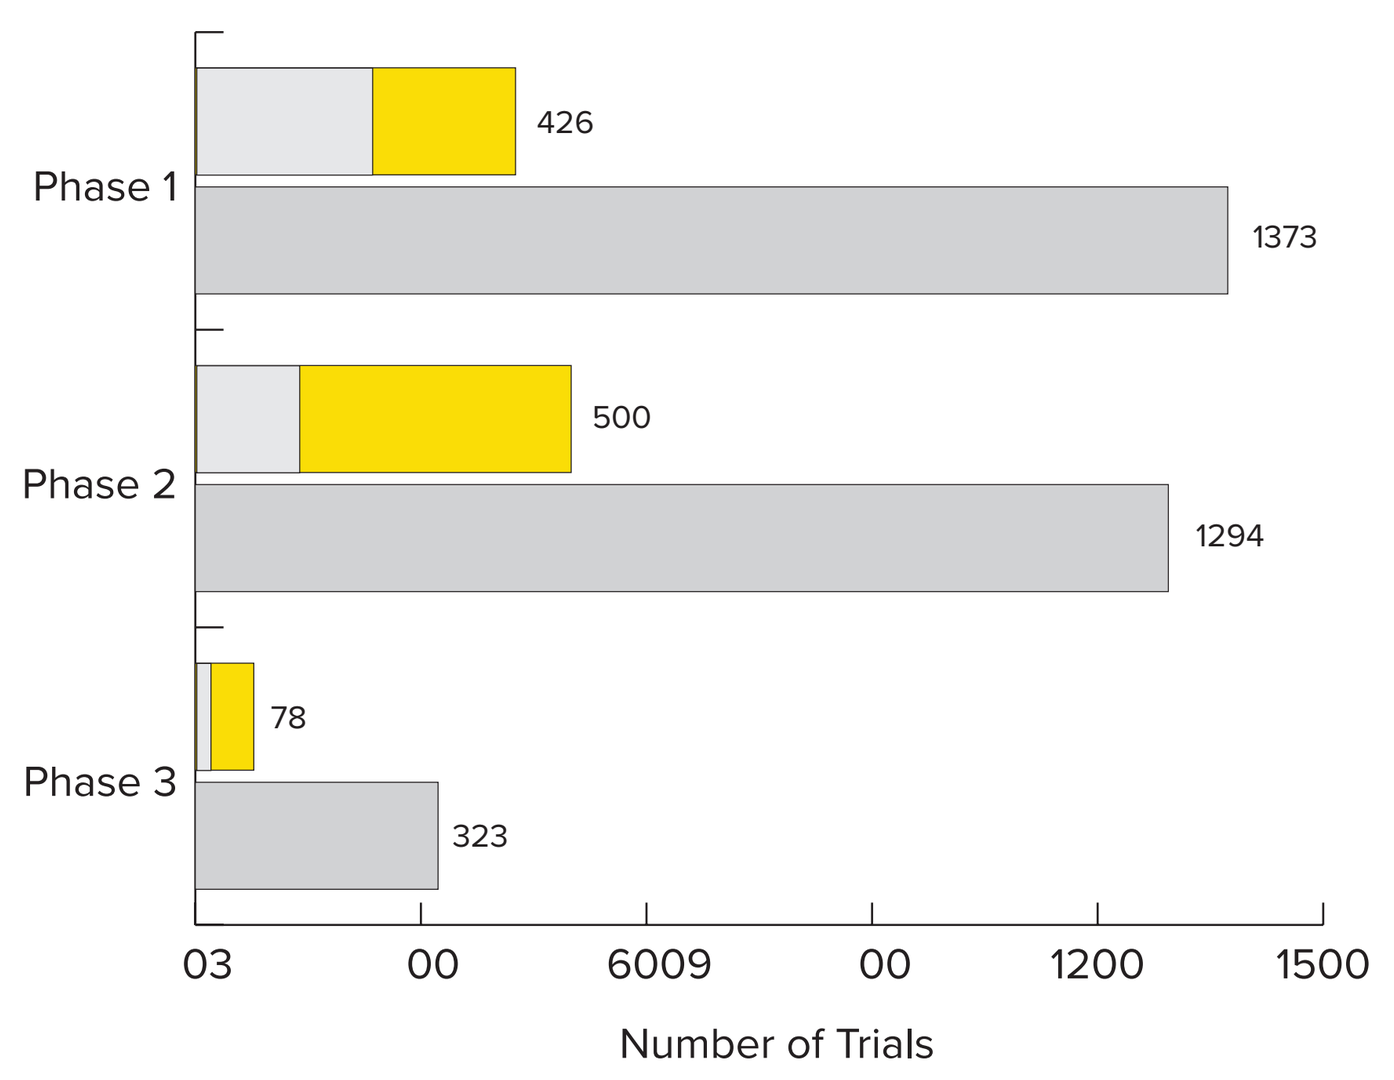
Phase 1
426
1373
Phase 2
500
1294
Phase 3
78
323
03 00 6009 00 1200 1500
Number of Trials
Rare trials
Mixed trials
Non-rare trials

Rare cancers were assessed using the ACS fewer than 6 per 100,000 metric (fig. 1).

FIGURE 2F: 74.89% OF PHASE 1-3 CLINICAL TRIALS DO NOT INCLUDE A RARE CANCER

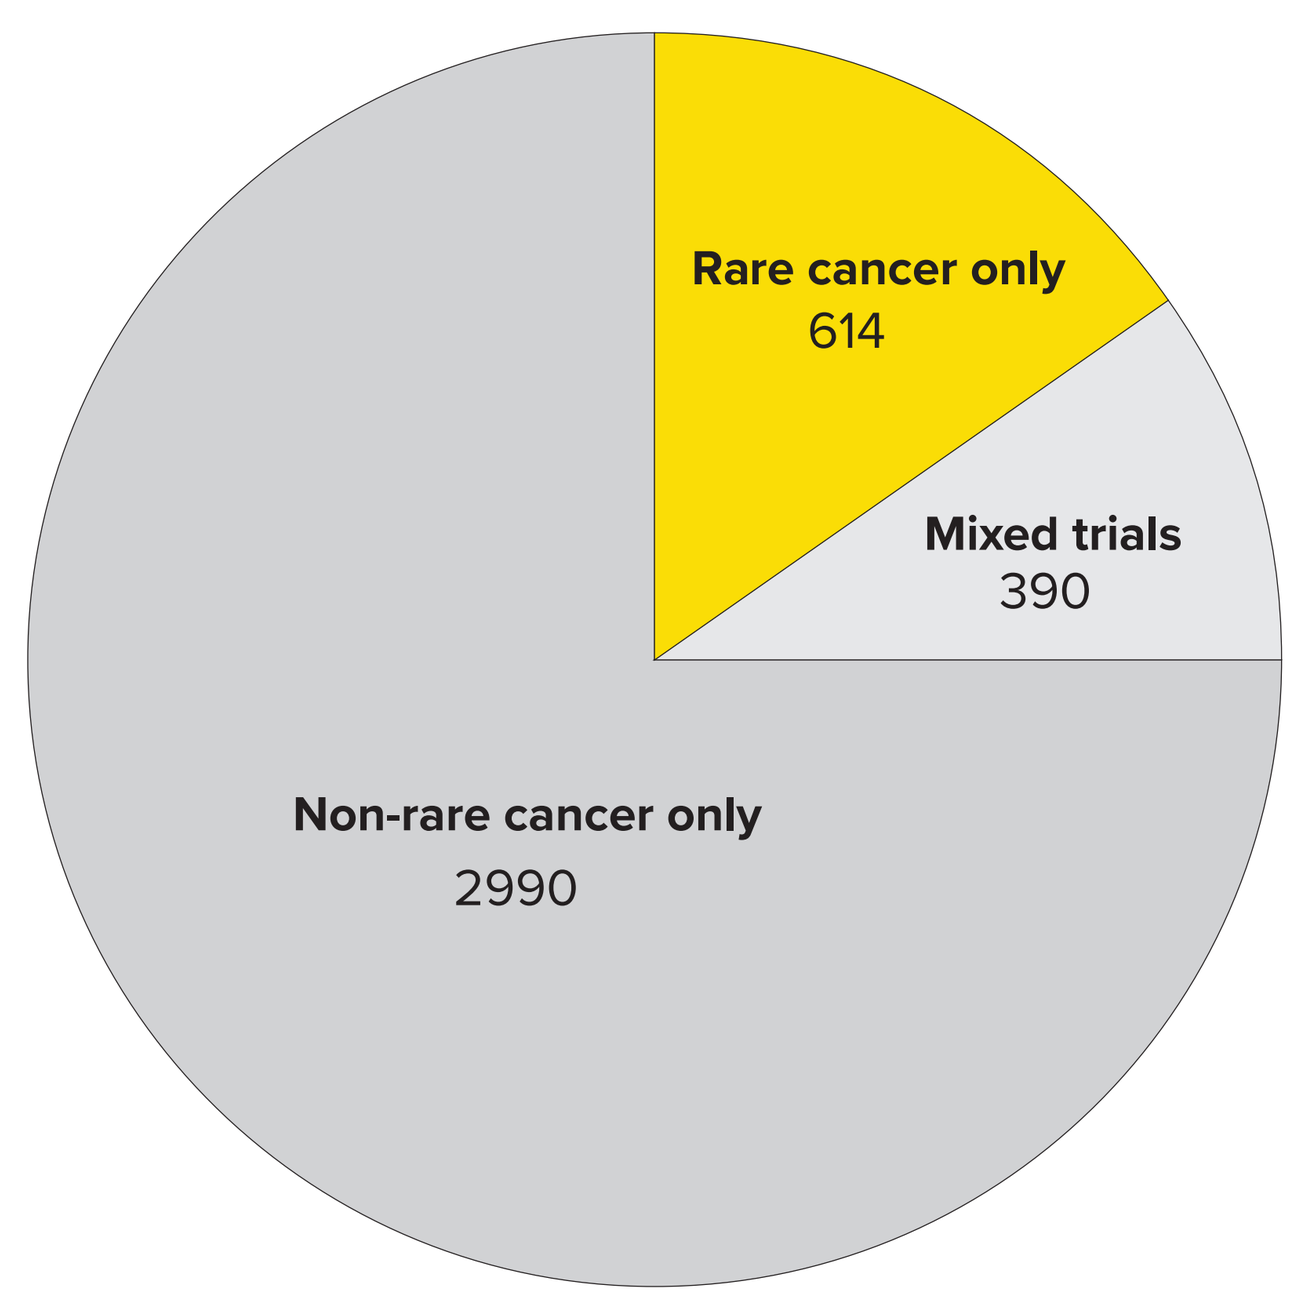
Rare cancer only
614
Mixed trials
390
Non-rare cancer only
2990

A total of 3994 oncology clinical trials in phases 1, 2, and 3 were identified between January 1, 2012 and January 1, 2017. Using the ACS fewer than 6 per 100,000 metric (fig. 1), of these trials, 2990, or 74.89%, did not include a rare cancer.

Clinical trials listed on clinicaltrials.gov were assessed from January 1, 2012 until January 1, 2017. 4309 total trials were accessed via clinicaltrials.gov. 318 trials were not assess because the trials involved intervention that was not designed as a specific cancer therapy, but rather studied conditions related to or caused by cancer (e.g. hair loss, nausea, etc.) 3994 total trials were assessed. The January 2012-December 2016 time frame was selected because it was the last full year for which data was available when this analysis was initiated. When incidence could not be determined for Phase III clinical trials, but the literature indicates it as extremely rare, we denote it as < 100 cases per year. Data collected from clinicaltrials.gov, a resource provided by the US National Library of Medicine.

FIGURE 3: PHASE 3 CLINICAL TRIAL PATIENT POPULATION AND SPENDING DISCREPANCY

FIGURE 3A: CLINICAL TRIAL COVERAGE FOR RARE AND NON-RARE CANCERS

	All cancers	Rare cancers (<6/100,000 incidence)	Non-rare cancers (>6/100,000 incidence)
Total indications in class	396	374	22
Cancers specifically named in phase 3 trials	72 (18.18%)	50 (13.8%)	22 (100%)
Cancers potentially covered by phase 3 trials due to subtyping	262 (66.16%)	242 (64.7%)	22 (100%)
Average 2017 incidence for specific cancers with trials	21,912	7,652	54,322
Estimated patients per year with access to a phase 3 trial	1,578,200 (93.26%)	388,673 (77.32%)	1,189,527 (100%)

FIGURE 3B: ESTIMATED PATIENTS PER YEAR WITHOUT ACCESS TO PHASE 3 DEVELOPMENT

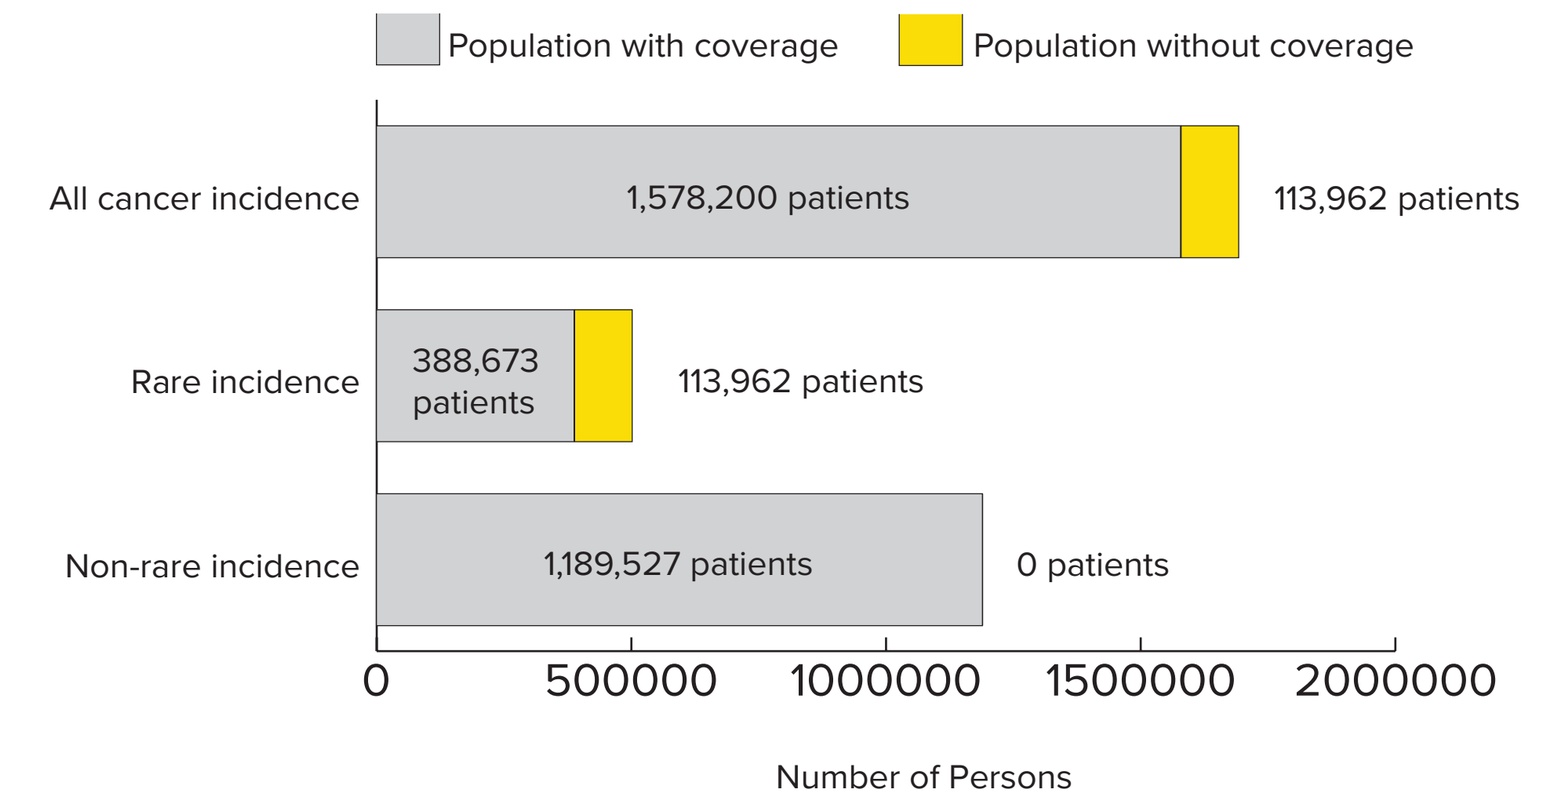
Population with coverage
Population without coverage
All cancer incidence
1,578,200 patients
113,962 patients
Rare incidence
388,673 patients
113,962 patients
Non-rare incidence
1,189,527 patients
0 patients
0 500000 1000000 1500000 2000000
Number of Persons

FIGURE 3C: ESTIMATED PHASE 3 CLINICAL TRIAL EXPENDITURE, 2012-2016

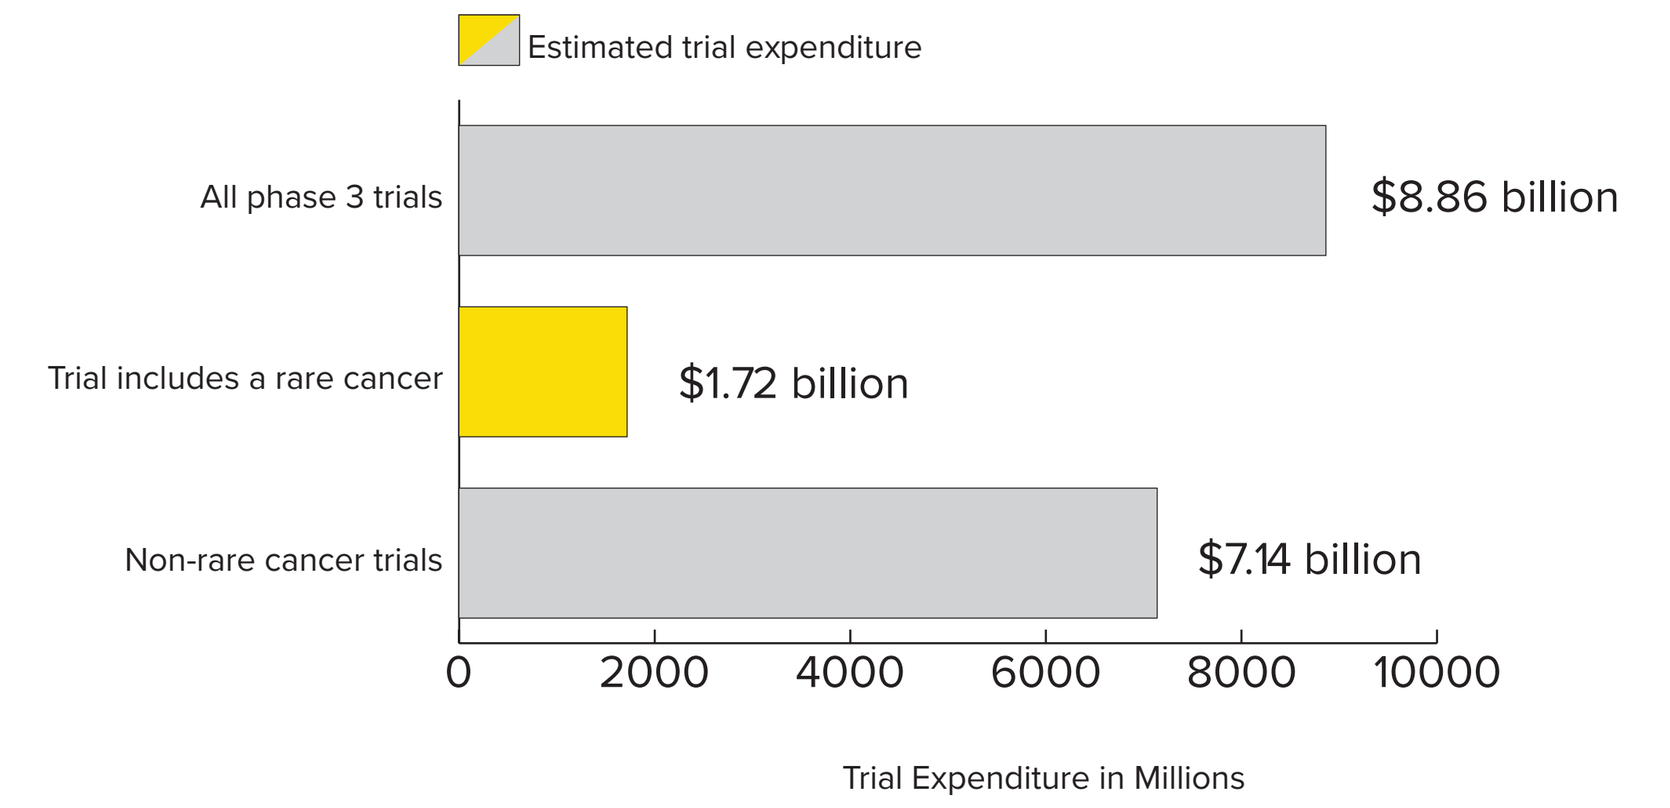
Estimated trial expenditure
All phase 3 trials
$8.86 billion
Trial includes a rare cancer
$1.72 billion
Non-rare cancer trials
$7.14 billion
0 2000 4000 6000 8000 10000
Trial Expenditure in Millions

72 different, specific cancers were listed a total of 426 times across 397 phase 3 clinical trials conducted between January 1, 2012 and January 1, 2017. Because several of these cancers have multiple forms, to honor conservatism, we assessed their potential audience across all indications in row 3 of figure 3A. Only the ACS metric was used to honor statistical conservatism, and to better focus on its implications. Average phase 3 clinical trial cost of $22.1 million was ascertained via Sertkaya, Aylin, et al. "Key cost drivers of pharmaceutical clinical trials in the United States." Clinical Trials 13.2 (2016): 117-126. Patients with access to a clinical trials is likely over-estimated due to molecular and pre-treatment exclusion criteria that cannot be assessed on an indication basis.

FIGURE 4: TOTAL PATIENTS WITHOUT AT LEAST ONE FDA-APPROVED TARGETED THERAPY

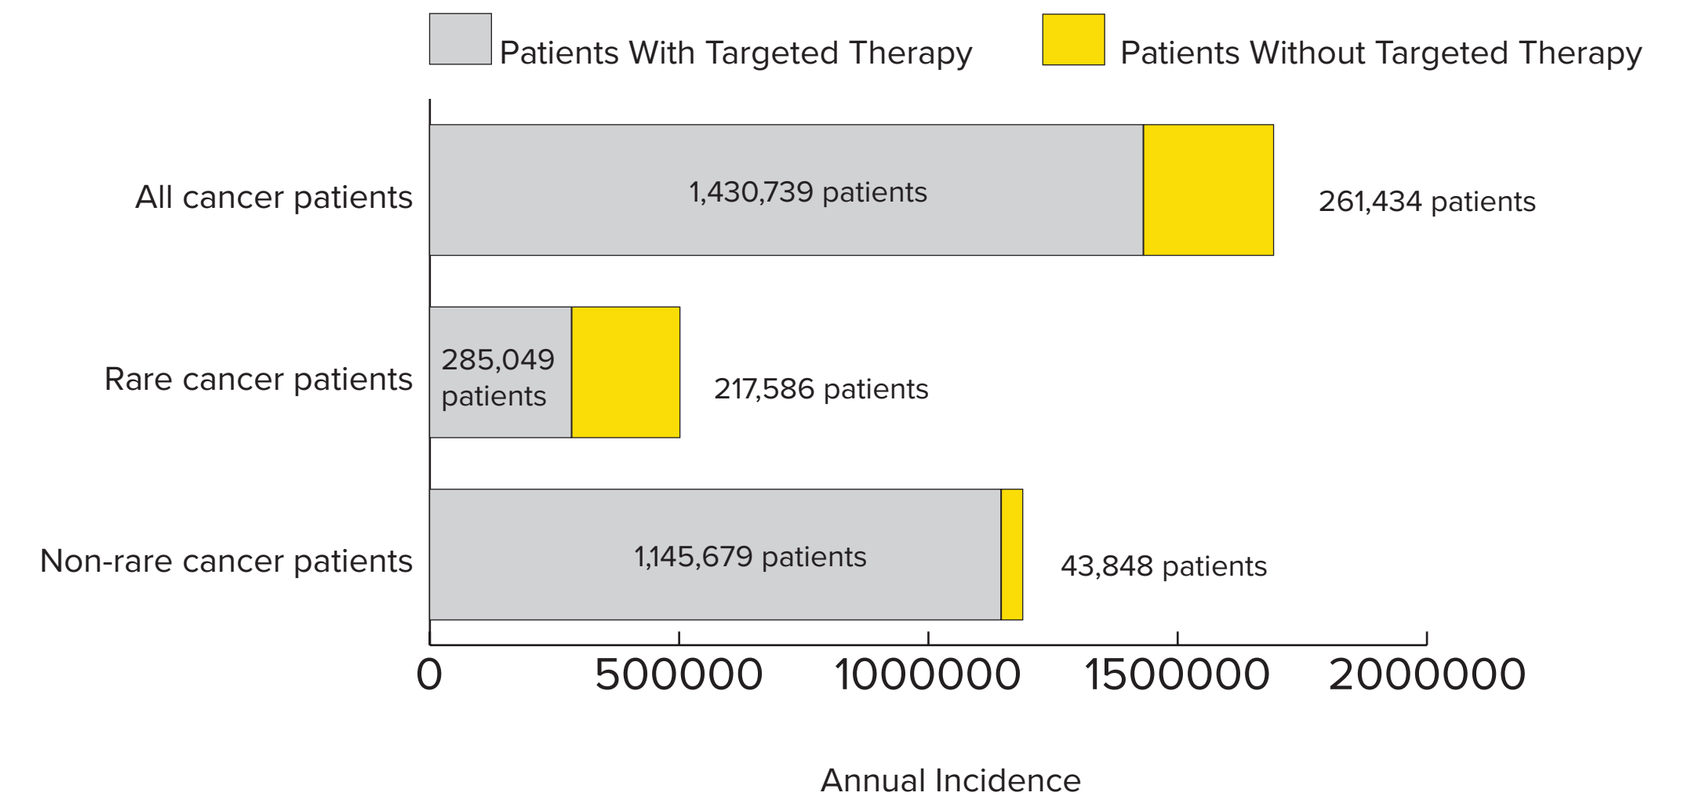
Patients With Targeted Therapy
Patients Without Targeted Therapy
All cancer patients
1,430,739 patients
261,434 patients
Rare cancer patients
285,049 patients
217,586 patients
Non-rare cancer patients
1,145,679 patients
43,848 patients
0 500000 1000000 1500000 2000000
Annual Incidence

The FDA had approved 92 targeted therapies for 56 indications as of March 1, 2018. Analysis of these indications conducted and applied to the rare cancer list. Classes of indication, such as breast, were assessed for the incidence of their rare and non-rare forms for the purposes of this analysis. Graphic is solely based on 2017 incidence population. This analysis does not consider molecular subtyping, survival benefit, or proportion of first-line and second-line therapies.

CONCLUSIONS & NEXT STEPS

I. Depending on the definition used, up to half of all rare cancer diagnoses in the US can be deemed rare. According to our conservative estimates using the ACS metric, nearly 30% of them are considered rare.

II. Across all clinical trial phases, rare cancers are disproportionally underserved, a particularly salient statistic at the gap between Phase 2 and Phase 3 clinical trials.

III. Even though the rare-cancer incidence could account for up to half of all cancer diagnoses in 2017 (by the NCI metric), 83.23% of patients who lack access to even one FDA-approved targeted therapeutic are rare cancer patients.

- Ideally, we would have preferred to categorize cancer subtypes according to molecular variables, but currently neither the literature nor principle databases come close to employing this strategy as the chief characterization variable. This highlights the need for a universal shift to using molecular data to characterize and categorize cancers.
- Our analysis demonstrates that the standard definitional metrics for "rare" have a statistically subjective basis lacking centrality in empirical approach. The trickle-down effect of economics and policies associated with this subjectivity is visible in the total phase 3 spending alloted to rare cancers, in the translation into therapies, and in the oversized incidence of rare cancers in marginalized groups who suffer from rare cancers at disproportionately excessive rate.
- The extremely significant lack of phase 3 clinical trials including rare cancers highlights the need for bridging the gap from the early-stage clinical into approved therapies.
- Additional analysis of FDA-approved targeted therapies for survival benefit, first-line versus second-line usage, and real-world prescription frequency per cancer is warranted.

REFERENCES

The ASCO Post. (2017, May 23). "One in Five Cancers Diagnosed in the United States Is a Rare Cancer." Retrieved January 1, 2018, from http://www.ascopost.com/News/55663?utm_medium=Email&utm_source=ExactTarget&utm_campaign=&utm_term=6643655.

Greenlee, R. T., Goodman, M. T., Lynch, C. F., Platz, C. E., Havener, L. A., & Howe, H. L. (2010). "The occurrence of rare cancers in US adults, 1995–2004." Public health reports, 125(1), 28-43.

"Targeted Cancer Therapies." National Cancer Institute. Accessed April 09, 2018. https://www.cancer.gov/about-cancer/treatment/types/targeted-therapies/targeted-therapies-fact-sheet#q4.

shepherd.bio Cambridge • Natick • Nashville